Acceptability of a new dose regimen for an oral solution of paromomycin after its dilution in milk or milk replacer in newborn calves

D. Achard[1], A. Trotel[1], G. Pagny[1], AG. Besnard[2]

[1]*Ceva Santé Animale, Libourne, France, damien.achard@ceva.com*
[2]*Cebiphar, Fondettes, France*

Objective

Cryptosporidiosis is a frequent parasitological infection in calves resulting in considerable economic losses. Current treatment options are limited. Paromomycin is considered as a valuable option and recently a new dose regimen has shown great results to cure calves sick from cryptosporidiosis (Gabbrovet Multi®, Ceva Santé Animale, 150 mg/kg of paromomycin sulfate per kg BW/day for 5 consecutive days via oral route). To ensure that young calves would accept this new dosage when mixed with whole milk (WM) or milk replacer (MR), a palatability study was performed with Gabbrovet Solution® (Ceva Santé Animale) as a positive comparator. Gabbrovet Solution® is indicated for colibacillosis and frequently used diluted in milk or milk replacer with no impact on feed intake at the maximum dosage of 50 mg/kg/day for 5 days.

Materials and methods

A GCP compliant study was performed in 30 newborn calves according to a blinded, randomized, and positive controlled design. At arrival calves were randomized into two feed regimens (WM or MR) with age and body weight as blocking factors. At day 0, calves aged between 4-14 days were enrolled if they present with no apparent sign of disease and a normal feeding behavior/appetite. Calves in each feeding group were randomly allocated to three study groups. Calves in group A (N=12) received 150 mg paromomycin sulfate/ kg b.w. (Gabbrovet Multi®), once daily, for 5 days diluted in 2L of WM or MR (morning feeding), calves in group B (N=12) received 50 mg paromomycin sulfate/ kg b.w. (Gabbrovet Solution®), once daily, for 5 days diluted in 2L of WM or MR (morning feeding), while sentinel calves in group C (N=6) received the same amount of WM or MR (2L) without any supplementation. The following acceptability and safety parameters were monitored for 5 days: feed intake, duration of the meal, health status. Comparison of the morning feed intake of calves for five minutes after meal distribution into bucket between calves in group A and B was the primary criteria to evaluate palatability.

Results

- Overall, intake values were close to 2L for all calves. Mean feed intake over the treatment period was 1948 ± 205 g in group A, 1902 ± 309 g in group B and 1997 ± 18 g in group C (Figure 1).
- Complete/uncomplete feed intake was found to be similar in group A and B according to a generalized mixed model ($p = 0.0752$).
- By study day, similar values were observed regarding meal duration scores and feed intakes, which highlights a regular acceptability over the treatment period.
- Calves managed to finish their supplemented meal within 5 minutes for 91.7 % of calves group A and 81.8% in group B in the whole D1-D5 period.
- Effect of the diluent (WM or MR) was not significant. This mode of administration was well tolerated as no adverse event were recorded to be related to the use of the product.

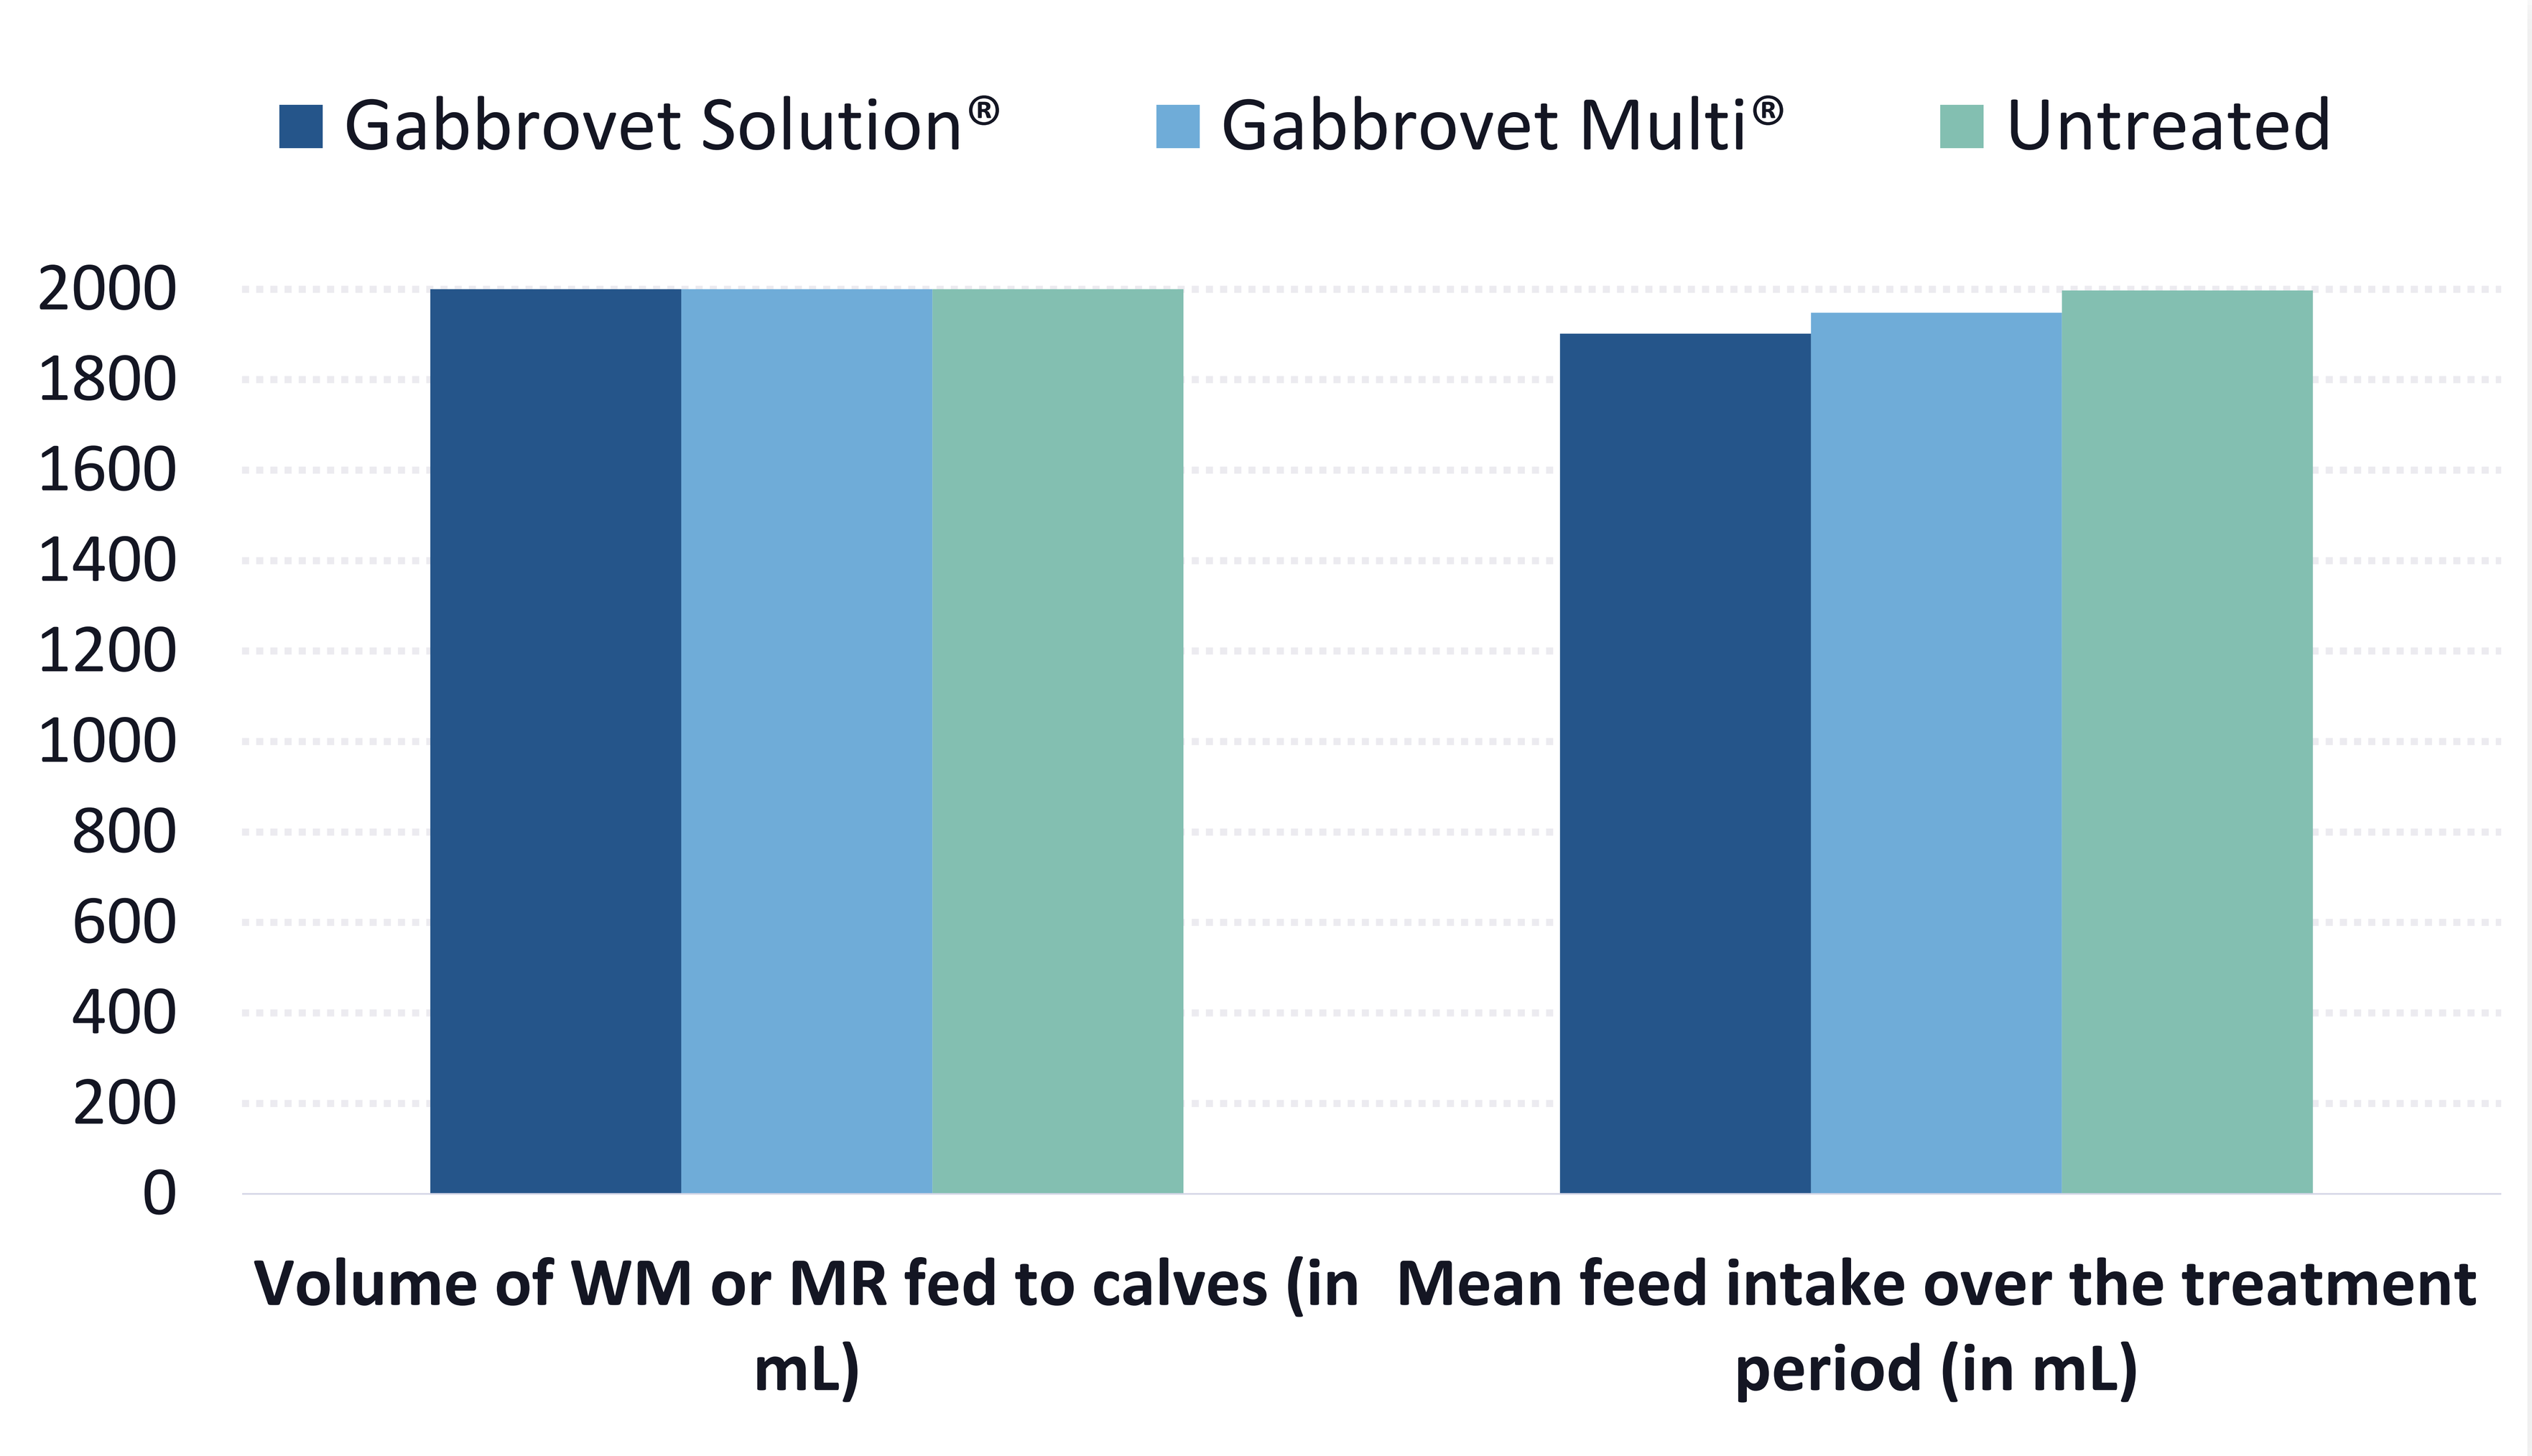
Gabbrovet Solution®
Gabbrovet Multi®
Untreated
2000
1800
1600
1400
1200
1000
800
600
400
200
0
Volume of WM or MR fed to calves (in mL)
Mean feed intake over the treatment period (in mL)

Figure 1. Feed intake in newborn calves fed with Gabbrovet Solution®, Gabbrovet Multi® or no supplementation diluted in WM or MR

Conclusions

- **In this experimental study, daily oral treatment with 150 mg/kg of paromomycin (Gabbrovet Multi®, Ceva Santé Animale) for 5 days was found to be safe and very well accepted by newborn calves when diluted in whole milk or milk replacer for their morning meal.**

References
Data on registration dossier (Study CLT/C651/1607): Evaluation of C651 palatability at the dose of 50 mg A97452/kg BW, administered to young calves orally in milk and milk replacer.